ENSEMBLE DISCOVERY

Programmed Macrocycle Libraries for Challenging Therapeutic Targets

Macrocyclic compound libraries are being created using DNA programmed chemistry and screened for activity against “extended binding motif” targets. The intramolecular ring closure, a challenging reaction via conventional chemical methods, and the introduction of side-chain diversity are both facilitated through a highly specific DNA-programmed reaction sequence. The libraries are assayed through highly sensitive affinity-based in vitro selections to identify small molecule ligands that bind to drug discovery targets of interest. Each compound carries a unique DNA tag, allowing measurement of the abundance of each compound before and after the selection process. Screening and analysis of the libraries is under way against targets relevant to the Oncology, Inflammation, and Anti-Viral therapeutic areas.

Overview

Ensemble Discovery uses DNA Programmed Chemistry (DPC) to generate libraries of programmed macrocycles as drug candidates to address targets with extended binding motifs.

Extended Binding Motifs: A Need for New Drug Modality

Extended Binding Motif

E.g.- Bcl-2 with BH3 peptide
Protein:Protein Interaction

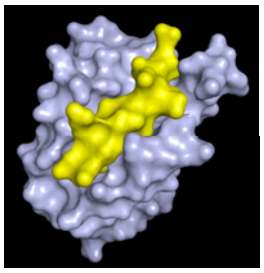

Defining Characteristics:
- Diversity of specific interactions
- Interfaces dispersed, complex
- Small-molecule hits rare

Discrete Binding Site

E.g.- Abl Kinase with inhibitor
Defined ATP Binding Site

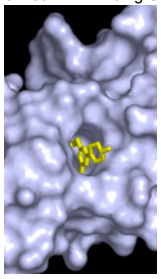

Defining Characteristics:
- Natural ligand is a small-molecule
- Well defined active site
- Small-molecule hits abundant

Macrocycles Suited for Modulation of Extended Binding Sites

Macrocycles: Important Properties

- Larger size and numerous distributed binding interactions
 - Enable recognition of extended binding motifs
- Amenable to incorporation of diversity
- Cyclic structure confers structural pre-organization
 - Reduced entropic loss on binding
 - Enhanced affinity compared with linear molecules (e.g. HCV protease inhibitors)
- Designed with pharmaceutical properties
 - Greater metabolic stability than linear molecules
 - Intramolecular H-bonds provide conformers with amphipathic properties (e.g. cyclosporin)
- Macrocyclic drug precedents
 - Cyclosporin, rapamycin, tacrolimus
 - Taxol, Taxotere, Integrilin, Protease inhibitors

Macrocyclic HCV protease inhibitor*

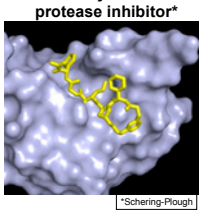

*Schering-Plough

Conformers of Cyclosporin A

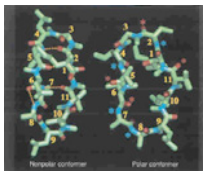

Nonpolar conformer
Polar conformer

Ensemble’s Control of Reactivity Enables Unique Macrocycle Capability

Ensemble Advantages
- **Ring closure in library format challenging with conventional chemistry; controlled through DPC**
- **Side chain diversity enabled by DPC**
- **Library synthesis includes purification and characterization**

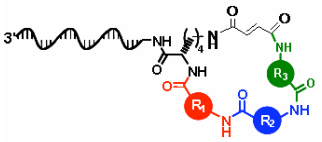

3'
NH
NH
R1
R2
R3

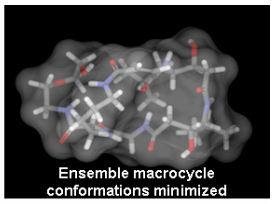

Ensemble macrocycle conformations minimized

Chemical Libraries

Each member has a unique DNA sequence attached to it

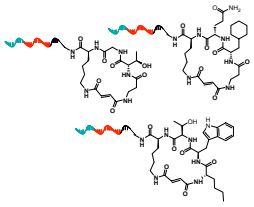

A library of DNA is prepared and small molecules are synthesized in a sequence dependent process. There is a one-to-one relationship between a DNA sequence and an individual library member. The DNA is subdivided into codons and each codon has a one-to-one relationship with an individual building block.

DNA tags provide a purification handle

Synthesis of DNA-encoded small molecule libraries includes a purification step to remove unreacted materials. Following macrocyclization, this purification ensures that only cyclized products are present in the final library.

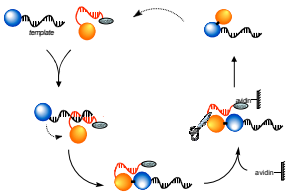

template
avidin

Rapid Interrogation of Diverse Chemical Libraries

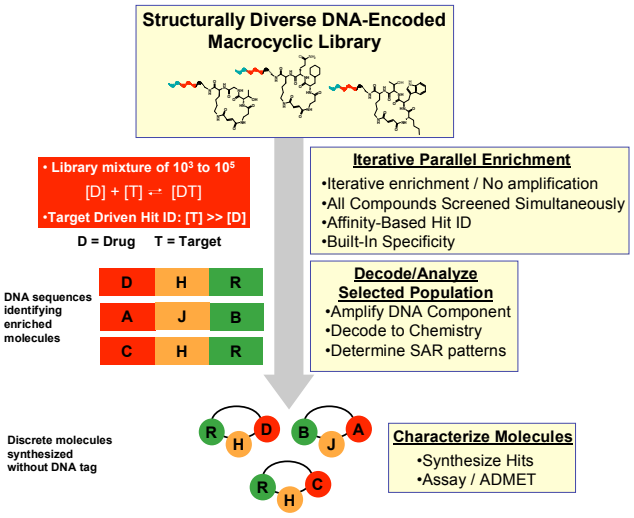

Structurally Diverse DNA-Encoded Macrocyclic Library
• Library mixture of 10³ to 10⁵
[D] + [T] ⇌ [DT]
•Target Driven Hit ID: [T] >> [D]
D = Drug T = Target
DNA sequences identifying enriched molecules
D H R
A J B
C H R
Discrete molecules synthesized without DNA tag
Iterative Parallel Enrichment
•Iterative enrichment / No amplification
•All Compounds Screened Simultaneously
•Affinity-Based Hit ID
•Built-In Specificity
Decode/Analyze Selected Population
•Amplify DNA Component
•Decode to Chemistry
•Determine SAR patterns
Characterize Molecules
•Synthesize Hits
•Assay / ADMET

Compound Recovery Based on User-Defined Parameters
A key advantage of the selection platform is the ability to interrogate collections of molecules simultaneously. The characteristics of the molecules selected are determined by user-defined parameters (Affinity, Specificity, Binding Site)

Visualization

Visual Display of Coded Molecules in a Cube

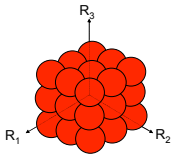

R3
R1
R2

We display libraries of compounds as points in a 3D structural space grid. The structural identity of each molecule defines its spatial positioning. The position along each of the three coordinates is a function of the chemical building blocks used to create that molecule.

Statistical Calculations

P-value calculations

We are interested in whether the distribution of members in a pool selected against an immobilized target differs from the distribution of those members in the starting pool (pre) or a pool selected against resin only (mock). We use a single tailed Fisher Exact test (Zhang 1998) to obtain the probability of the null hypothesis, that two of the samples are drawn from an identical distribution.

Multiple Hypothesis Correction

Using a p-value cutoff of 0.05 would result in 1000 false positives from a 20,000 member library. Instead, we tolerate 20% of the members that pass our cutoff as being false, which corresponds to using a false discovery rate (FDR) with an alpha being equal to 20%.

Our estimated error rate for a given sequence is: $\alpha*R/(C_N*N)$ where N is the size of the library and R is the sequence’s rank based on p value. C_N is Sum(1/i) for i=1 to N, used because the selection is dependent.

The cube can be filtered by p-value and enrichment

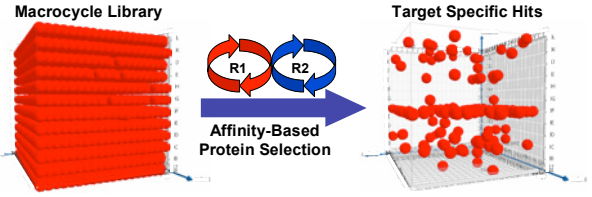

Macrocycle Library
Target Specific Hits
R1
R2
Affinity-Based Protein Selection

SAR against different target classes

Active building block combinations are observed against several targets

A library of 19,200 programmed macrocycles was selected against various target classes. 3D plots of significantly enriched sequences show patterns of planes (one active building block) and lines (combination of two active building blocks). Assays of discrete compounds corresponding to these library members are in progress.

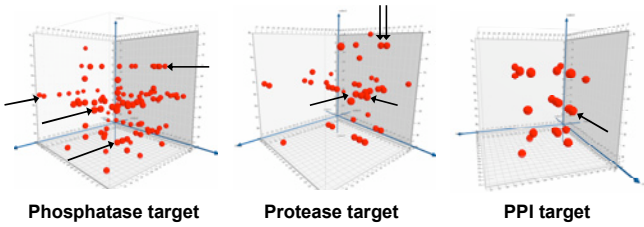

Phosphatase target
Protease target
PPI target

Conclusions

Ensemble Discovery uses DNA Programmed Chemistry (DPC) to generate high quality libraries of programmed macrocycles. These molecules have several potential advantages against traditionally difficult classes of protein targets. The unique features of DPC enable the synthesis of libraries that are difficult to access through traditional methods. Through a rapid selection and analysis process, molecules of interest can be identified as potential binders and further improved through focused secondary libraries. We have identified hit compounds and SAR patterns against a protease, a phosphatase, and a protein-protein interaction target.

Contact Information

Josh Bittker
Principal Scientist
jbittker@ensemblediscovery.com

Ensemble Discovery Corp
99 Erie St
Cambridge, MA 02139